DocuSign Envelope ID: F3A435D7-A74D-4CB3-9436-76D8DF582382

7254

EVALUATION OF scFv AND DIABODY PSEUDOTYPED SINDBIS ENGINEERED RETROVECTORS FOR TARGETED CANCER THERAPY

Makoto Sato[1], Alexander Cunha[1], Sara Collins[2], Noriyuki Kasahara[2], Cecilia Roh[1], and Robert G. Johnson[1]

[1]GenVivo Inc., San Marino, CA; [2]UCSF, San Francisco, CA

ABSTRACT

Engineering viral vectors to target specific cells and organs after intravenous delivery remains a challenge. Transcriptional targeting with cell-specific promoters is a commonly used approach but does not change vector biodistribution. We have developed a novel gene delivery system targeted to tumor cells by pseudotyping the vector with Sindbis Virus (SB) envelope bearing an antigen-binding domain. Prototype vectors were generated by inserting a universal IgG binding domain from Protein A into the SB envelope (ZZ SINDBIS). Antibodies (Abs) against various cell-surface targets (HLA, HER2, CD47, EGFR, Nectin-4), were conjugated to SB-ZZ vectors delivering a luciferase (Luc) reporter gene. Targeted transduction was confirmed on cells expressing the corresponding antigens by significantly higher Luc activity compared to SB-ZZ vectors without Abs. SB envelopes were further engineered by adding or removing furin cleavage sites and linkers, transduction-enhancing point mutations, and direct incorporation of single-chain variable fragment (scFv) in different VH and VL orientations, or diabody (DB) sequences, against the same targets. The specificity of optimized HER2-targeting vectors delivering the Luc reporter was evaluated in SKBR3 (HER2 over-expressing) and MDA-MB-468 (HER2-null) cells. Robust Luc activity was observed in SKBR3 cells, but only negligible activity in MDA-MB-468 cells. In MDA-MB-468 cells overexpressing HER2, significantly higher transduction was observed compared to the wild-type cells. When comparing HER2-targeting vectors with scFv vs. DB sequences based on the same antibody, transduction capability was higher with the DB motif in HER2-positive cell lines. Targeting vectors delivering HSV-eTK were also constructed, and ganciclovir (GCV)-mediated cell killing was evaluated *in vitro*. SKBR3 cells transduced with HER2-targeting vectors showed significantly more cell killing after GCV treatment than after transduction with an amphotropic reference vector. Our results indicate a method for directing gene therapy vectors to specific populations of tumor cells and provide a framework for highly modular targeted gene therapy and immunotherapeutics. Experiments are underway to examine *in vivo* delivery of targeted vs amphotropic vectors.

INTRODUCTION

GenVivo (GVO) retroviral vectors, delivering either Luc or copGFP (cGFP) as a reporter gene or Herpes simplex virus-derived enhanced thymidine kinase (HSV-eTK) as a suicide gene, were first pseudotyped with SB envelope (env) incorporating a Protein A-derived IgG-binding sequence insert (ZZ SINDBIS) with multiple mutations (GVO2.2), and evaluated for targeted transduction with multiple full-length Abs, including anti-HLA, anti-HER2, anti-CD47, anti-EGFR and anti-Nectin-4 Abs. We then further engineered the GVO2.2 env by restoring a furin cleavage site (GVO2.2F; Fig. 1), or by direct incorporation of either scFv (2nd Generation) or DB (3rd Generation) binding motifs (Fig. 2) by replacing the protein A sequence in the E2 subunit (Fig. 1). Examples shown in this presentation are anti-HER2 scFv (GEN331L) and anti-HER2 DB (GEN311L).

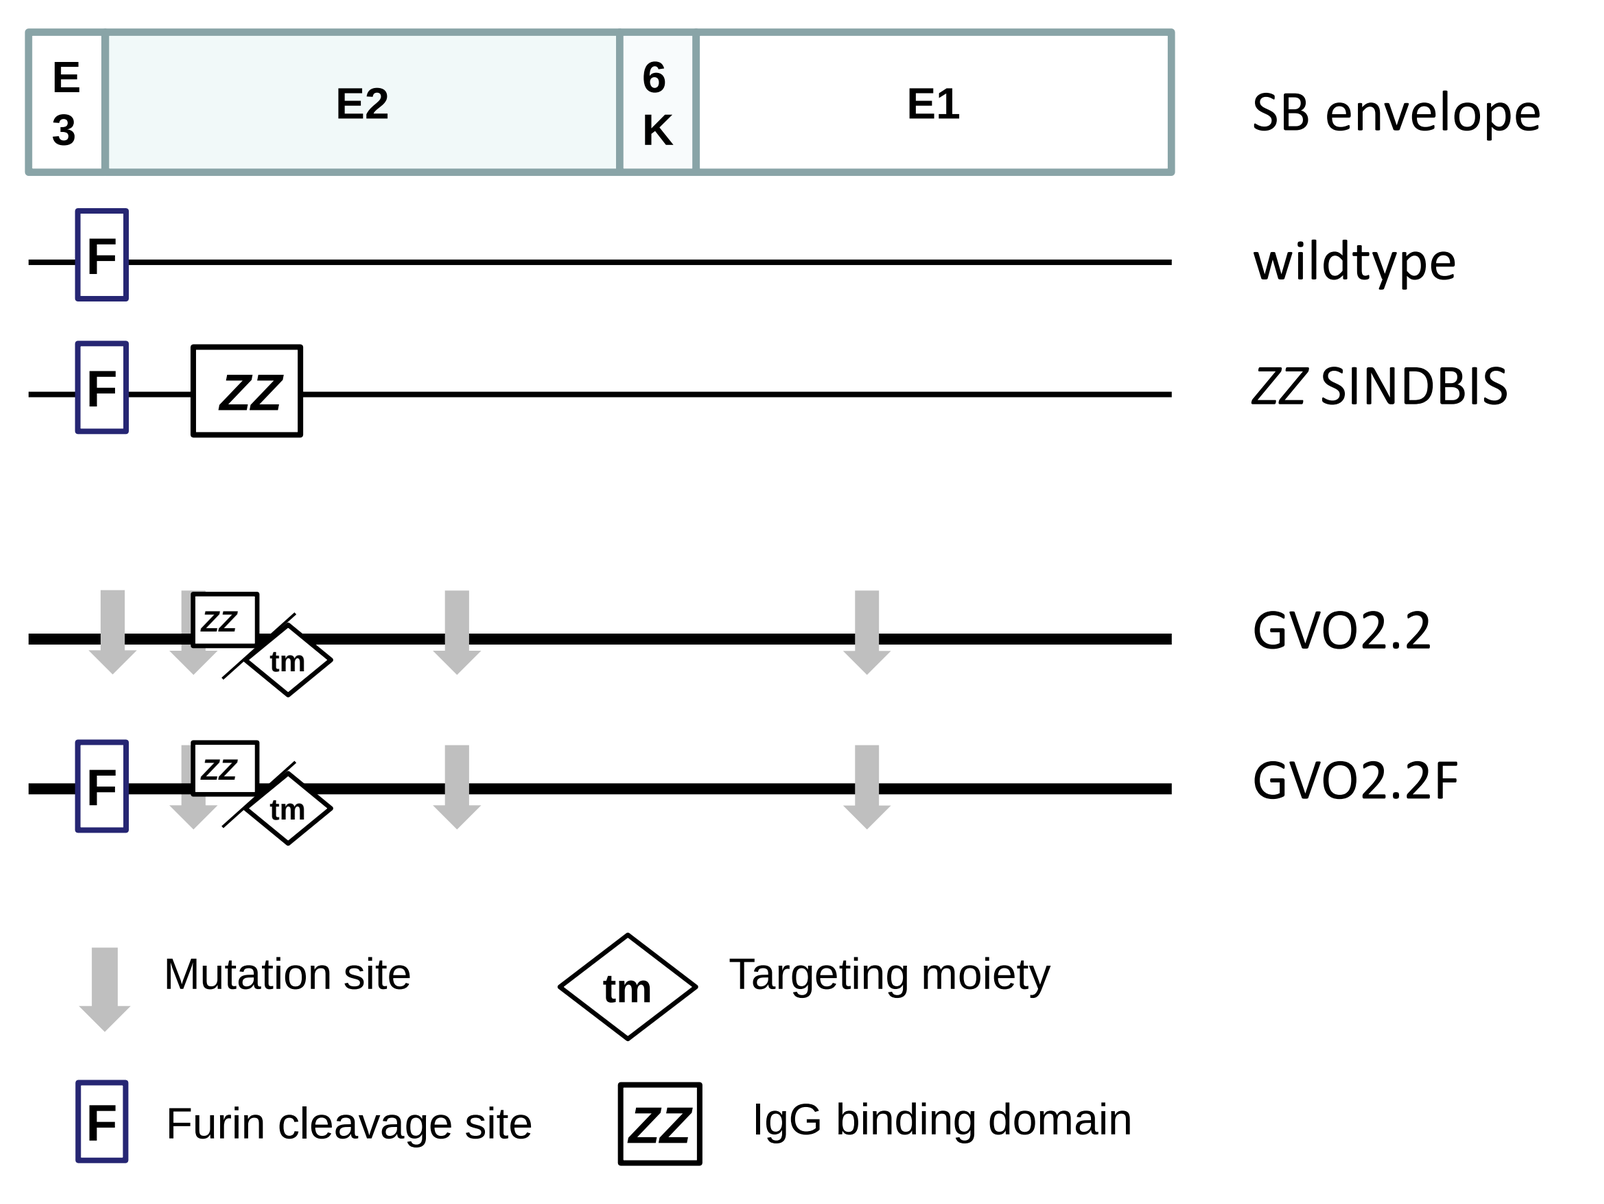
E3
E2
6K
E1
SB envelope
F
wildtype
ZZ
ZZ SINDBIS
tm
GVO2.2
GVO2.2F
Mutation site
Targeting moiety
Furin cleavage site
IgG binding domain

Fig. 1 Schematic of SB envelope for pseudotyping

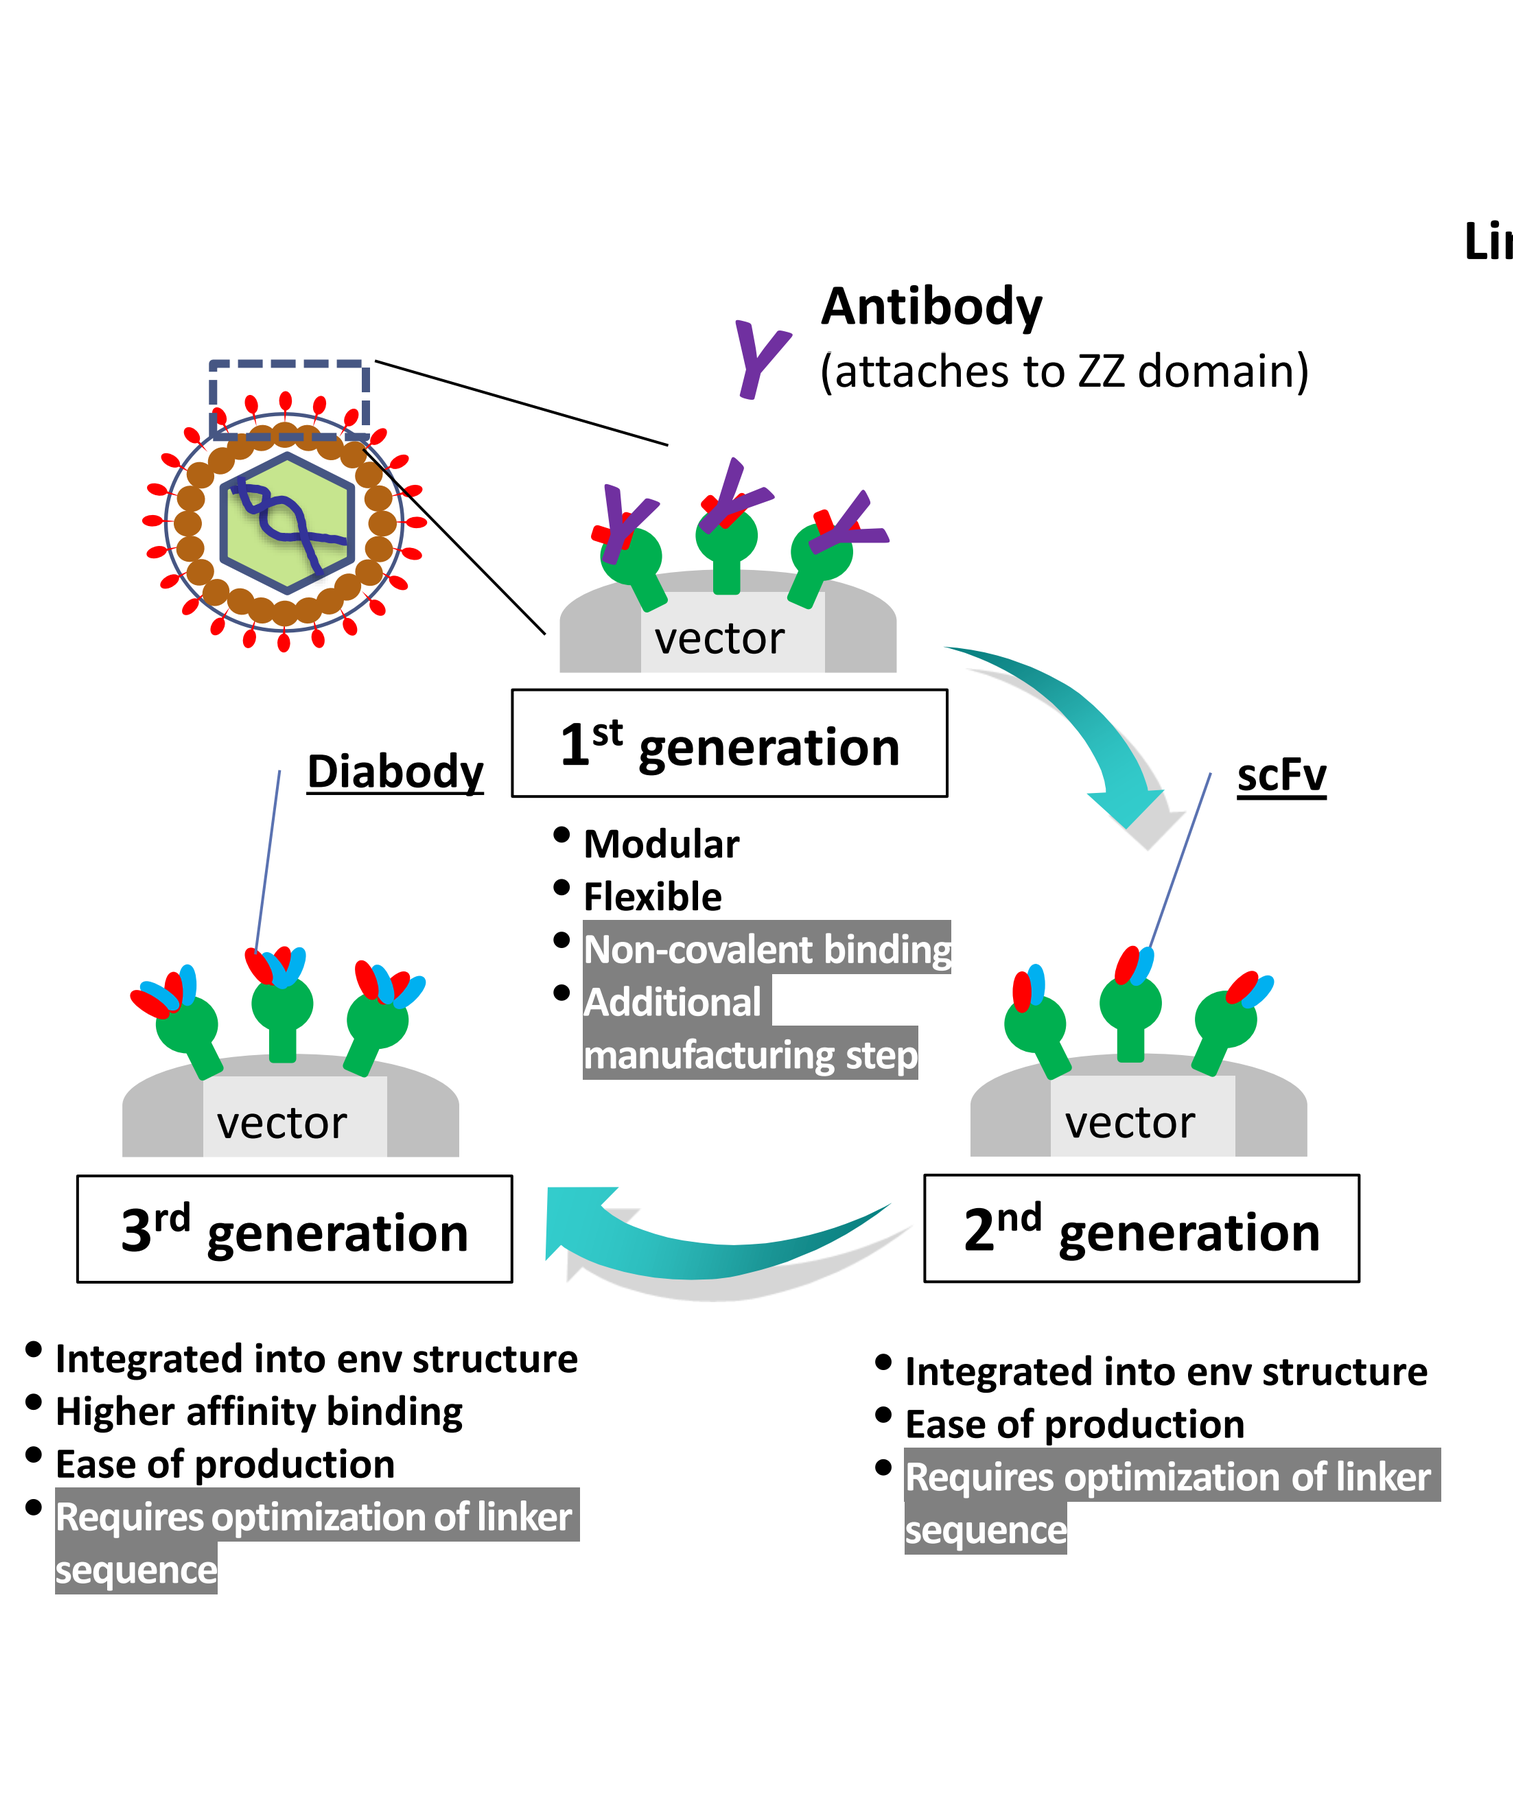
Antibody
(attaches to ZZ domain)
vector
1st generation
Modular
Flexible
Non-covalent binding
Additional manufacturing step
Diabody
scFv
3rd generation
Integrated into env structure
Higher affinity binding
Ease of production
Requires optimization of linker sequence
2nd generation
Integrated into env structure
Ease of production
Requires optimization of linker sequence

Fig. 2 Design strategies for targeted GVO vectors

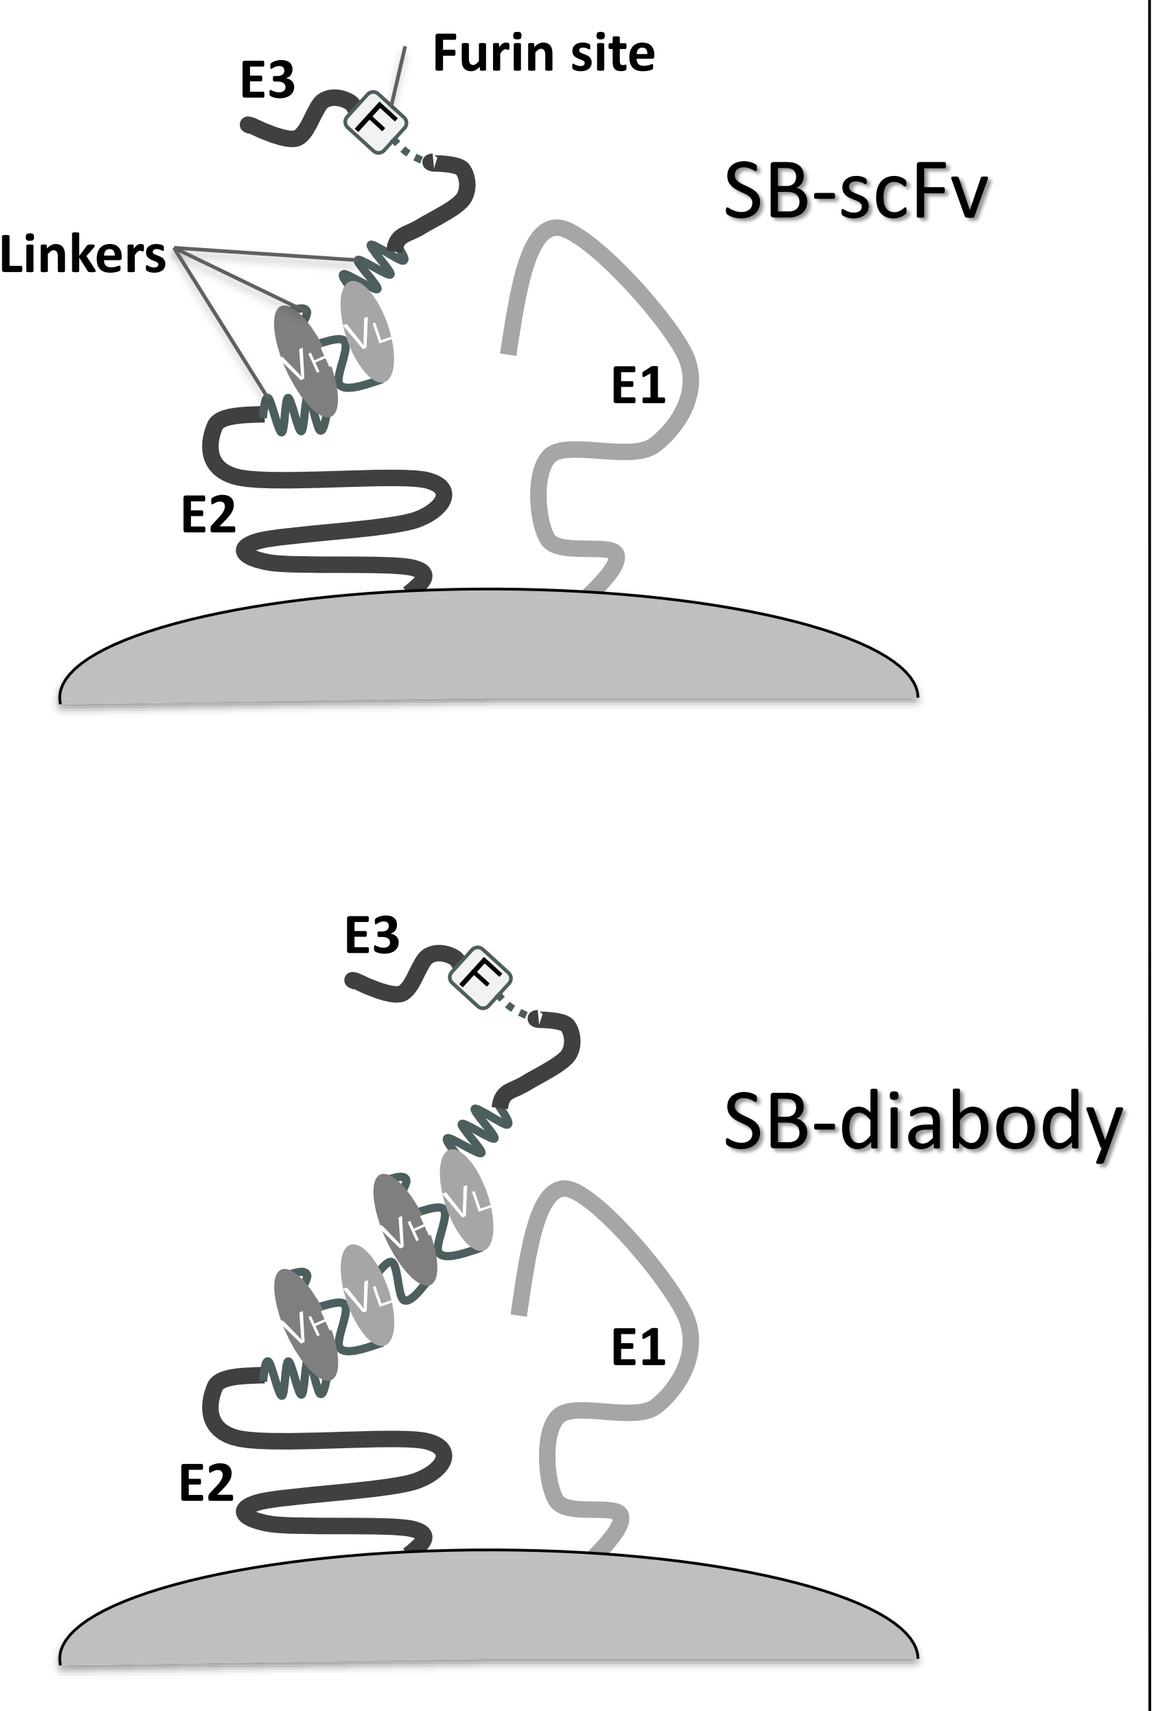
E3
Furin site
Linkers
SB-scFv
E1
E2
SB-diabody

Fig. 3 SB envelope with two different targeting moieties

METHODS AND MATERIALS

Constructs and Vectors Basic SB env genes were synthesized at GenScript and further engineered in-house to generate GVO2.2 or GVO2.2F constructs. ScFv and DB sequences were also synthesized at GenScript and cloned into BstEII sites of GVO2.2 or GVO2.2F constructs. All final plasmids were prepared at scale by GenScript and underwent whole plasmid sequencing at Primordium Labs. All vectors were produced by transient triple transfection to 293T cells with plasmids encoding gag-pol, payload, and envelope, according to GVO standard operating procedures (SOPs). For Ab conjugation, GVO2.2 and GVO2.2F-pseudotyped vectors were incubated with HLA-Ab (Invitrogen) or HER2-Ab (R&D Systems) for 15 minutes. For animal studies, vectors were purified and concentrated by the GenVivo Protein Science Engineering (PSE) team, and vector titers were measured by RT-qPCR according to GVO SOPs.

	Furin site (E3)	Targeting moiety	Transgene
Amphotropic			
RVE-Luc			Luc
GEN1017			cGFP
Sindbis-pseudotyped			
GVO2.2-Luc	–	ZZ (protein A)	Luc
GVO-SB-scFv-H-0-Luc	–	F+ α-HER2-scFv	Luc
GVO-SB-scFv-H-1-Luc	–	α-HER2-scFv	Luc
GEN331L	+	α-HER2-scFv	Luc
GVO-SB-DB-H-0-Luc	–	F+ α-HER2-DB	Luc
GVO-SB-DB-H-1-Luc	–	α-HER2-DB	Luc
GEN3111	+	α-HER2-DB	HSV-eTK
GEN311L	+	α-HER2-DB	Luc
GEN3117	+	α-HER2-DB	cGFP

Table 1 Vector nomenclature
Control retrovectors are pseudotyped with amphotropic env (4070A), which binds to a phosphate transport protein (Pit-2) on cell surfaces.
SB-pseudotyped scFv and DB vectors were constructed with (H-0) or without (H-1) a cryptic furin cleavage site inside the targeting moiety sequence (also indicated as “F+”).
α-HER2: anti-HER2.

Cell Culture and Transduction Studies Various cancer cell lines (ATCC) were cultured per GVO SOPs, and transduction studies were performed *in vitro* in the presence of 8 μg/mL polybrene. Three days later, transduced cells were analyzed by luciferase assay (Luc reporter) or flow cytometry (cGFP reporter). For cell-killing assays, cells were transduced with serially diluted HSV-eTK vectors and incubated with 20 μM GCV for 3 days. PrestoBlue was used to determine GCV-mediated cell-killing activity.

Animal Studies GEN3117 and GEN1017 were tested for *in vivo* transduction efficiency in a peritoneal carcinomatosis model using a firefly Luc-marked HER2-positive human ovarian cancer cell line (SKOV3ip-fLuc2). Five days after injecting SKOV3ip-fLuc2 cells intraperitoneally, 3e5 TU of each vector or buffer control was injected 4 times every 5 days. On day 23, animals were sacrificed and analyzed for vector transduction by flow cytometry.

RESULTS

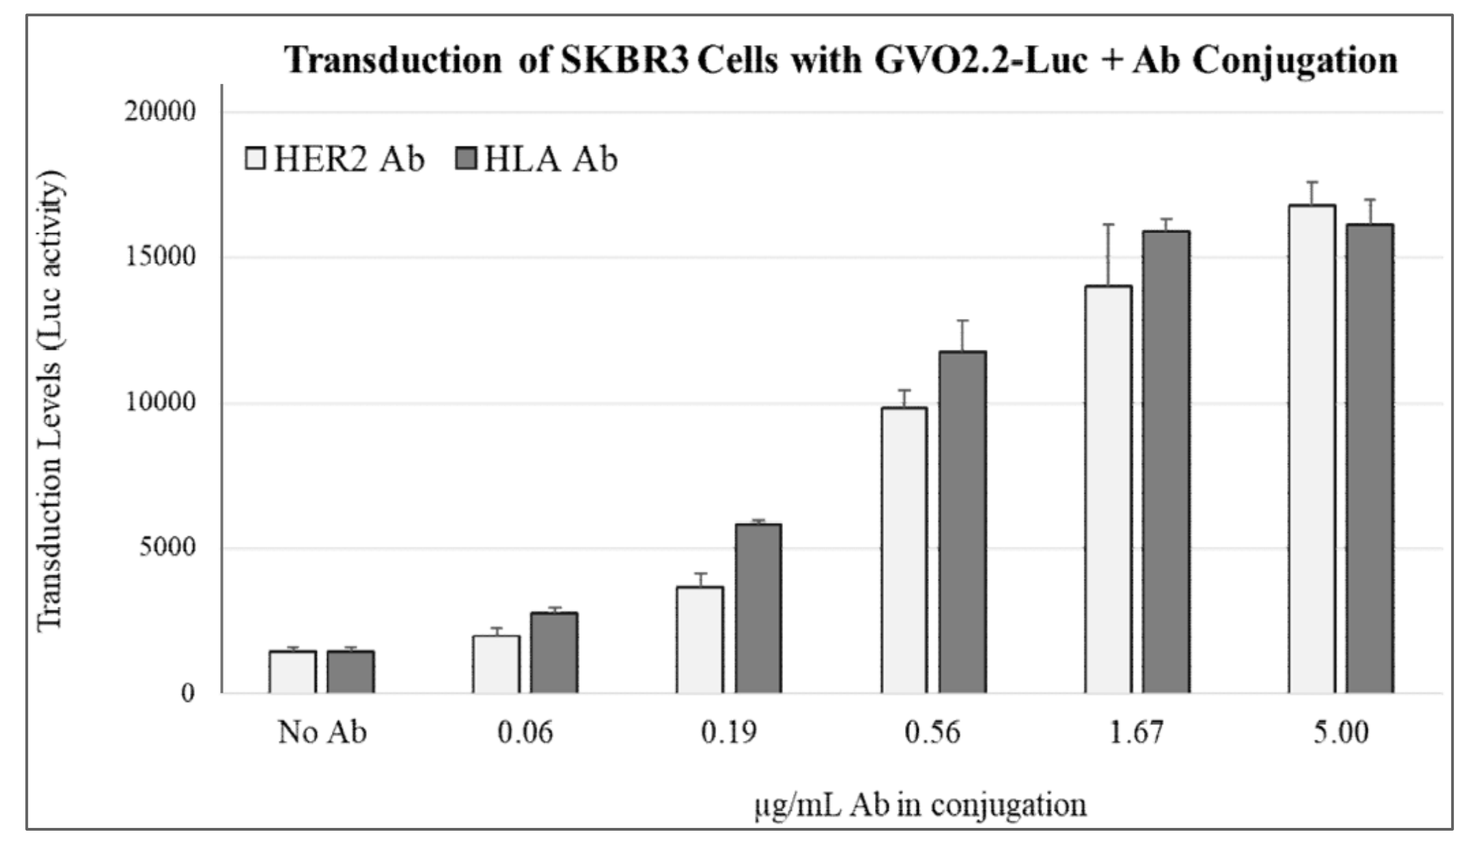
Transduction of SKBR3 Cells with GVO2.2-Luc + Ab Conjugation
HER2 Ab
HLA Ab
Transduction Levels (Luc activity)
No Ab
0.06
0.19
0.56
1.67
5.00
μg/mL Ab in conjugation

Fig. 4 Targeted transduction of SKBR3 cells by GVO2.2-Luc vector conjugated with anti-HER2 or anti-HLA monoclonal antibodies
HER2-overexpressing SKBR3 human breast cancer cells were incubated with anti-HLA or anti-HER2 antibody-conjugated GVO2.2-Luc vector prepared with different antibody concentrations. Three days post transduction, cells were assayed for luciferase activity. Transduction was seen with both Abs in an antibody concentration-dependent manner. HLA was used as a positive control for human cell line transduction, and HLA-Ab conjugated vector did not transduce human colon cancer HCT-15 cells, which lack HLA expression (data not shown).

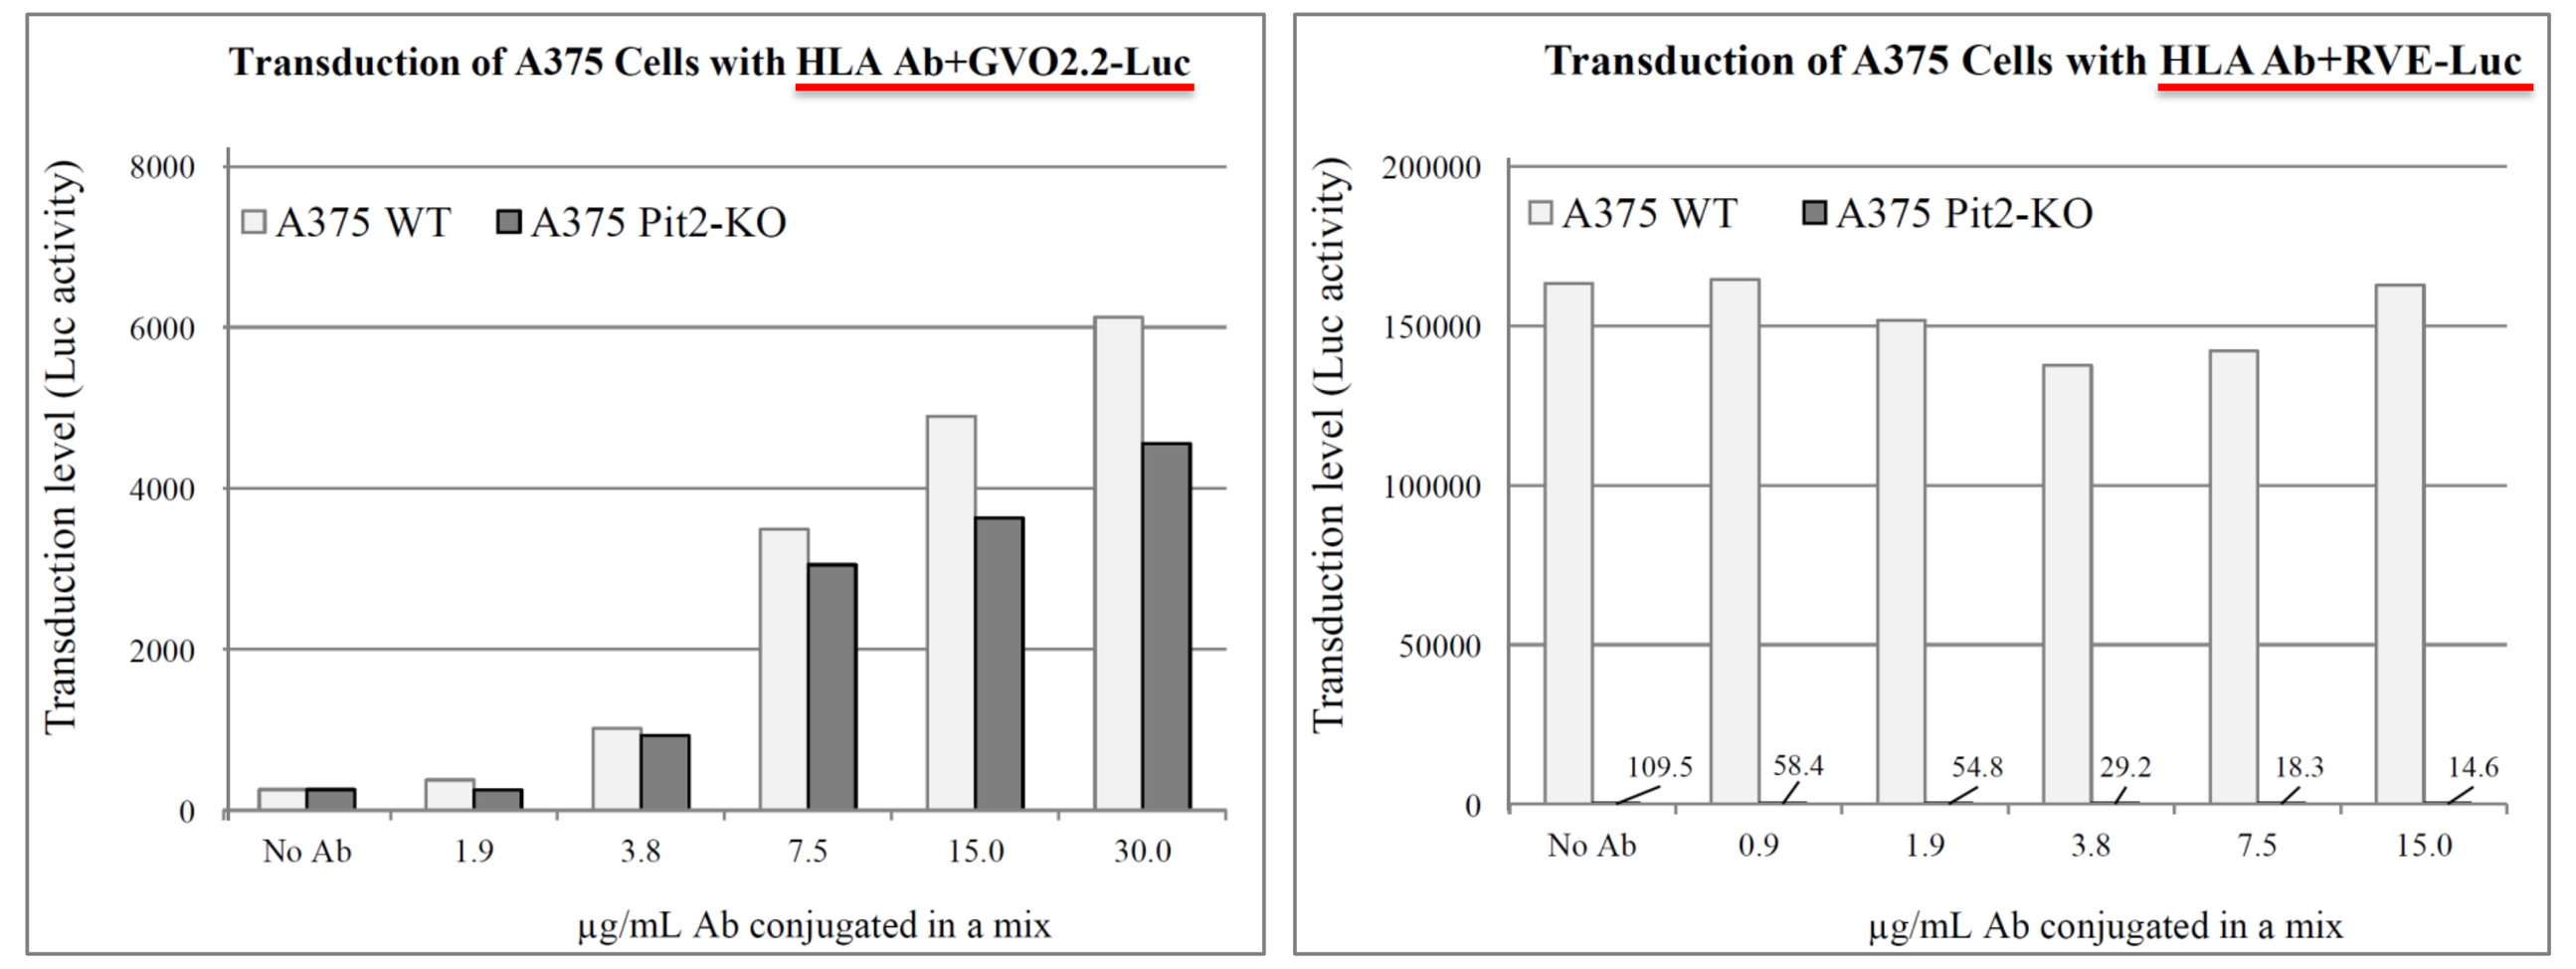
Transduction of A375 Cells with HLA Ab+GVO2.2-Luc
A375 WT
A375 Pit2-KO
Transduction level (Luc activity)
No Ab
1.9
3.8
7.5
15.0
30.0
μg/mL Ab conjugated in a mix
Transduction of A375 Cells with HLA Ab+RVE-Luc
109.5
58.4
54.8
29.2
18.3
14.6
No Ab
0.9
1.9
3.8
7.5
15.0

Fig. 5 Transduction of A375 wt and Pit2-Knockout (KO)-A375 cells by GVO2.2-Luc and RVE-Luc vectors conjugated with anti-HLA Ab
GVO2.2-Luc was conjugated with increasing concentrations of anti-HLA-Ab and incubated with wildtype or Pit2-KO A375 cells (generated in-house). HLA-Ab+GVO2.2-Luc vector successfully transduced both, while amphotropic control RVE-Luc vector failed to transduce Pit2-KO A375 cells even after addition of anti-HLA-Ab.

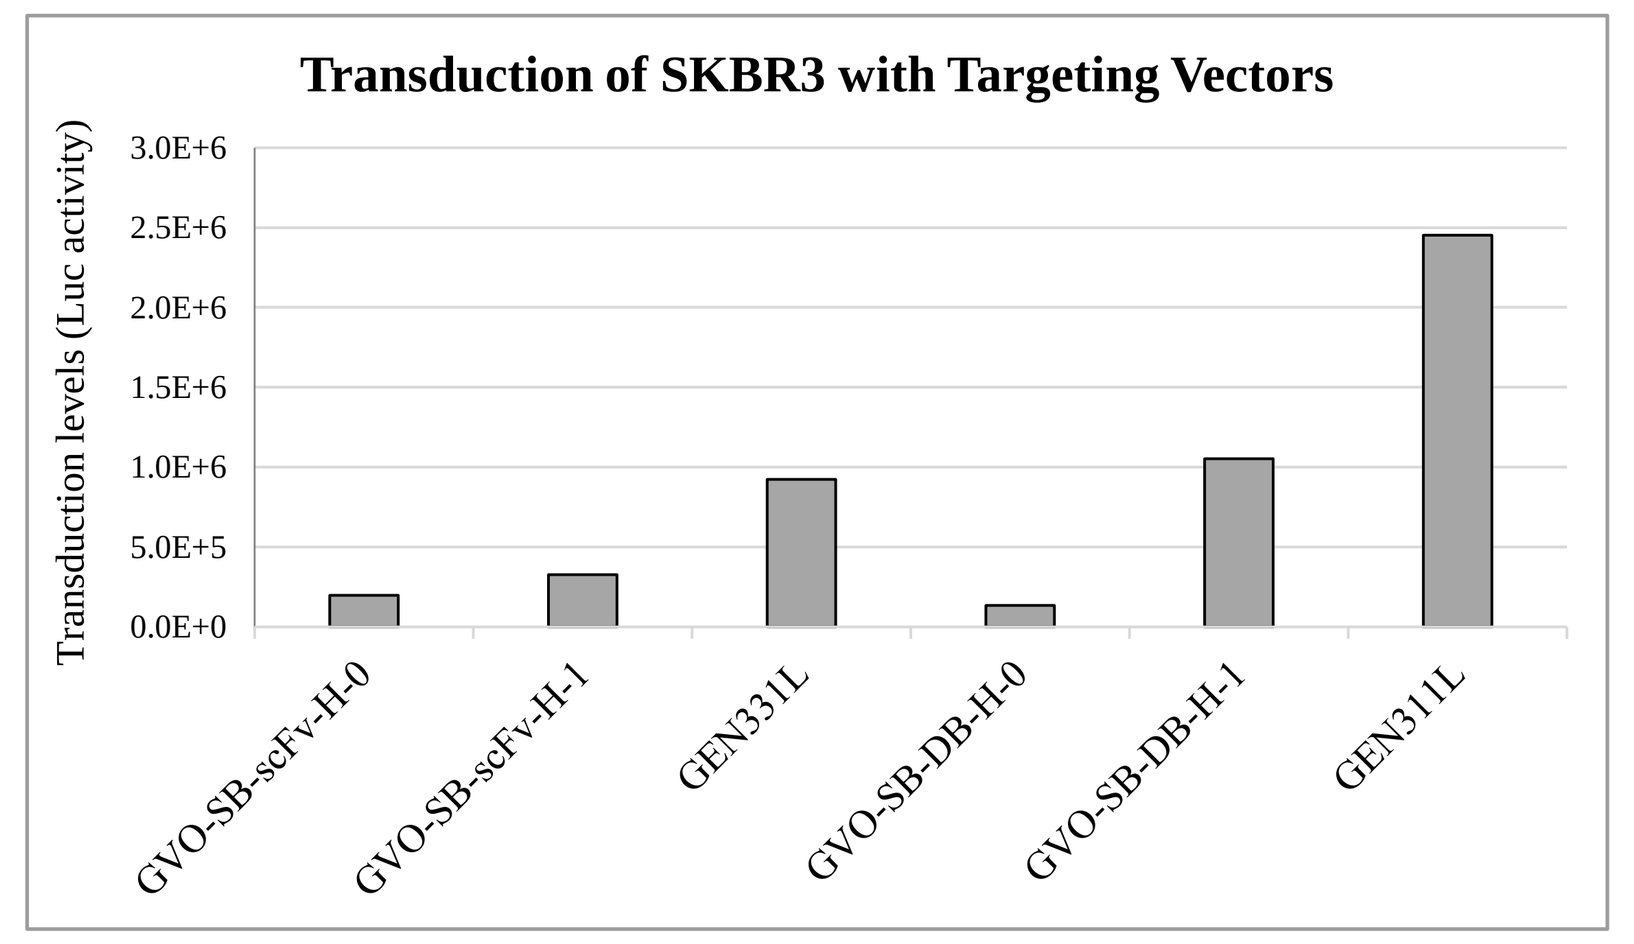
Transduction of SKBR3 with Targeting Vectors
Transduction levels (Luc activity)
GVO-SB-scFv-H-0
GVO-SB-scFv-H-1
GEN331L
GVO-SB-DB-H-0
GVO-SB-DB-H-1
GEN311L

Fig. 6 Sequence optimization to improve transduction levels
SKBR3 cells were transduced with HER2-targeting vectors with different backbone configurations. Left three: anti-HER2 scFv-carrying targeting vectors. Right three: anti-HER2 diabody(DB)-carrying targeting vectors. (**See Table 1**)

- Eliminating a cryptic furin cleavage site within the target-binding region greatly restored transduction as compared to the vector containing the cleavage site.
- However, placing the site between E3 and E2 significantly improved transduction.

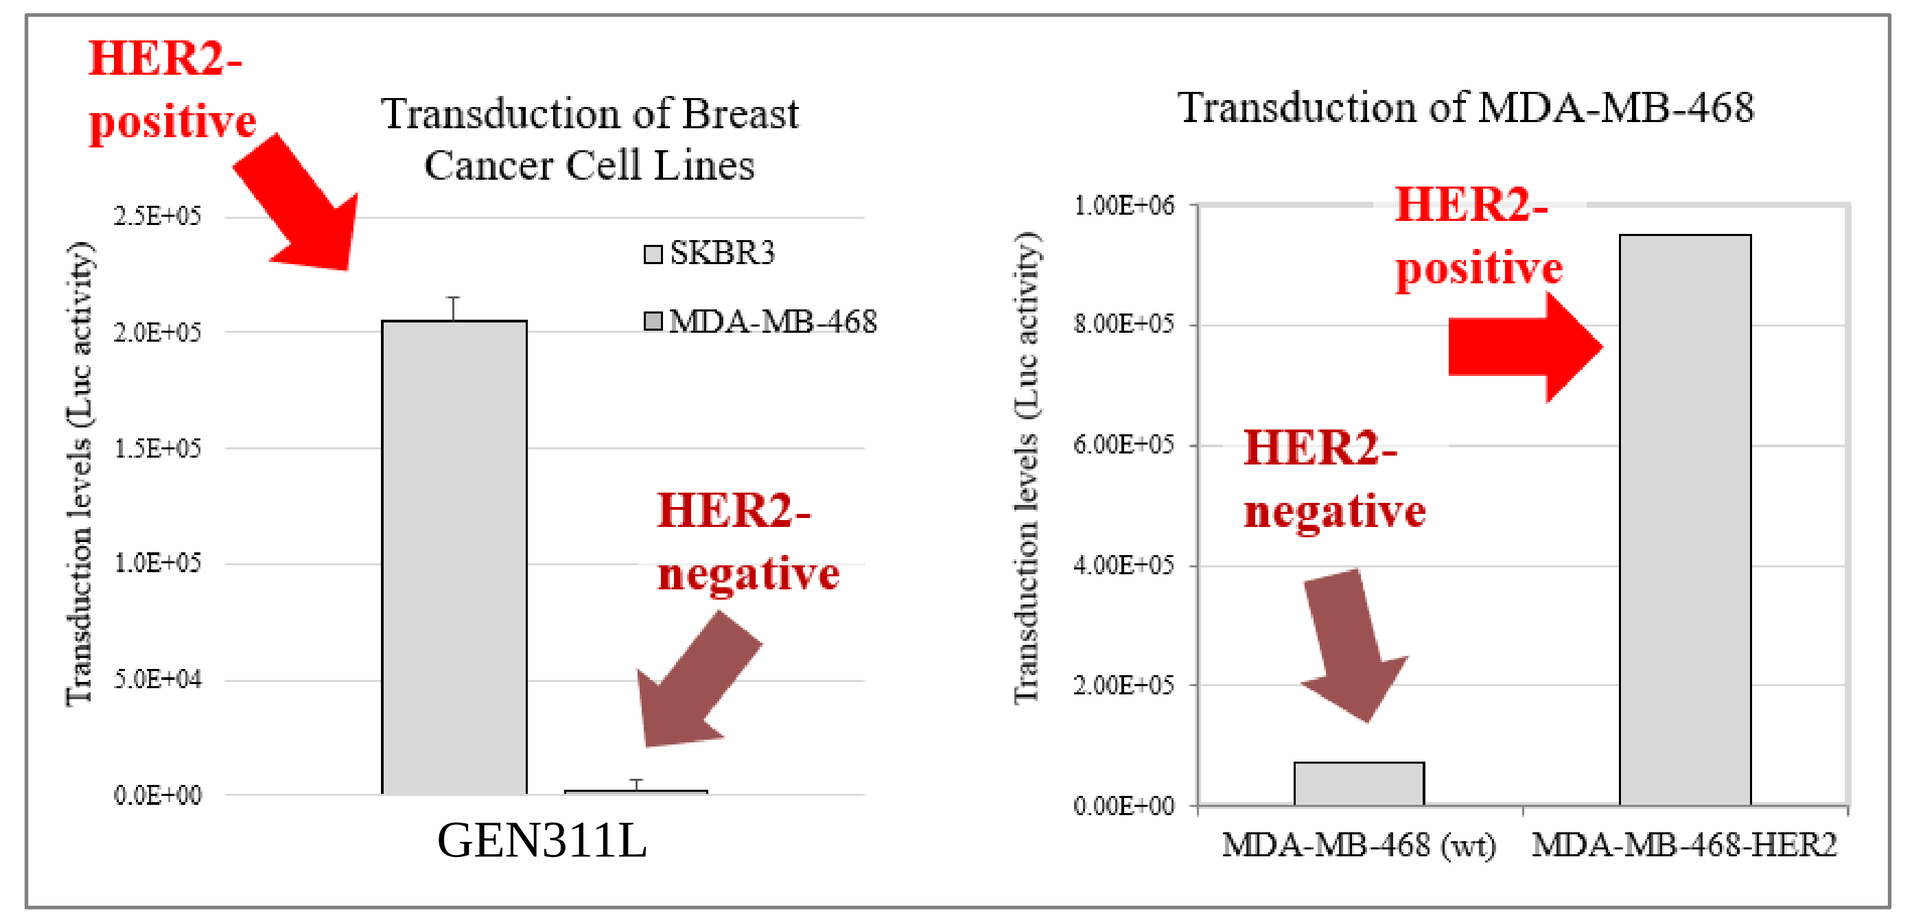
HER2-positive
Transduction of Breast Cancer Cell Lines
SKBR3
MDA-MB-468
HER2-negative
GEN311L
Transduction of MDA-MB-468
MDA-MB-468 (wt)
MDA-MB-468-HER2
Transduction levels (Luc activity)

Fig. 7 Specificity of GEN311L in HER2-positive vs. negative breast cancer cell lines
GEN311L vector transduces SKBR3 HER2-overexpressing cancer cells, (left panel) but not HER2-negative MDA-MB-468 cancer cells (right panel). However, expression of HER2 in MDA-MB-468 cells (generated in-house) rendered them susceptible to GEN311L (right panel). The HER2-targeting vector also transduced SKOV3 human ovarian cancer cells. In addition, transduction by HER2-targeting vectors was partially inhibited when cells were preincubated with HER-2 antibodies (data not shown).

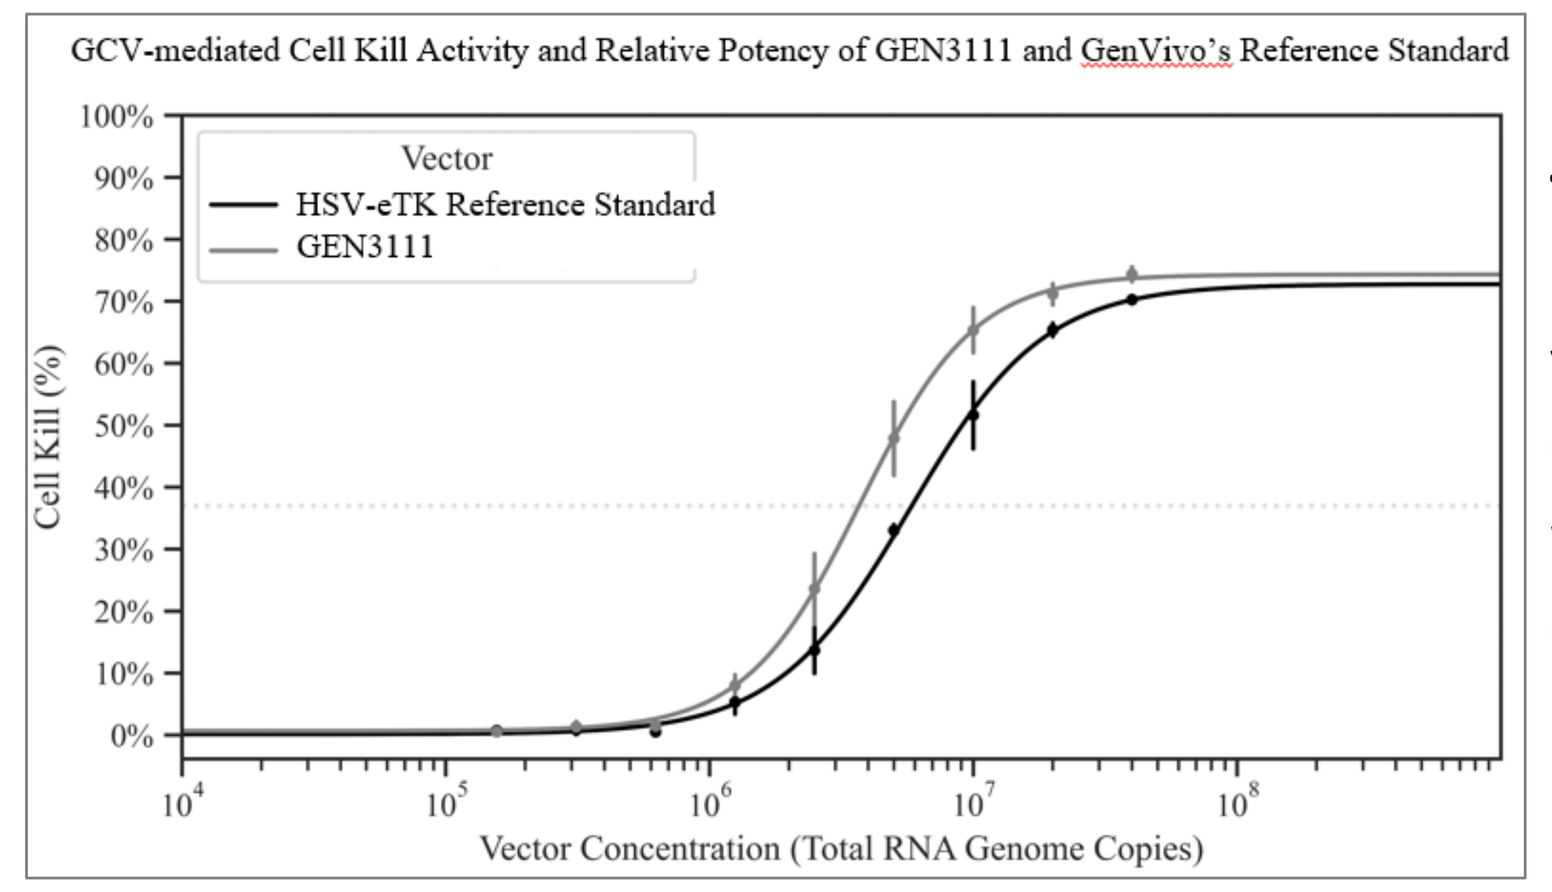
GCV-mediated Cell Kill Activity and Relative Potency of GEN3111 and GenVivo's Reference Standard
Vector
HSV-eTK Reference Standard
GEN3111
Cell Kill (%)
Vector Concentration (Total RNA Genome Copies)

Fig. 8 GCV-mediated cell-killing activity of HER2-targeting vectors using HSV-eTK as transgene
HER2-positive SKBR3 cells were transduced with serial dilutions of HER2-targeting vector (GEN3111) or amphotropic env-pseudotyped HSV-eTK vector. Next day, 20 μM GCV was added, and cells were cultured for three days. Using PrestoBlue, cell viability was measured by plate reader. The anti-HER2 DB targeted vector GEN3111 exhibited comparable to slightly higher cell killing activity than the control vector.

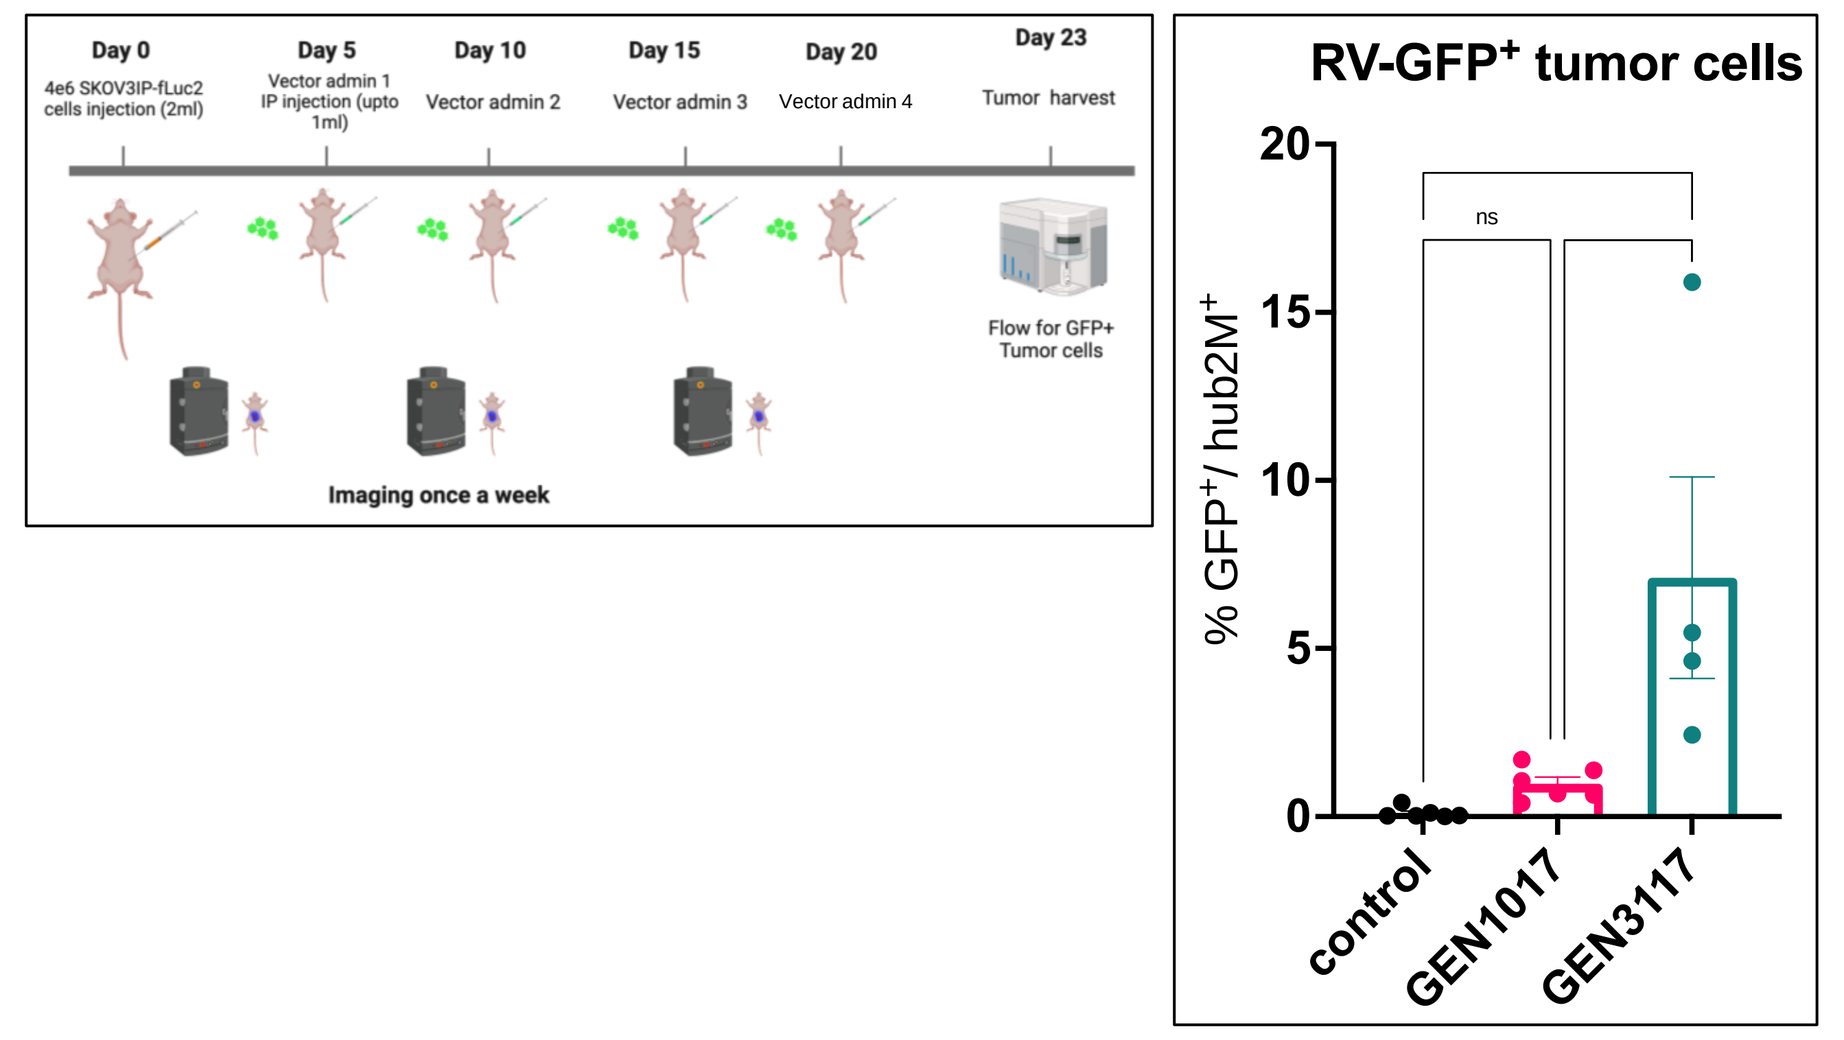
Day 0
Day 5
Day 10
Day 15
Day 20
Day 23
Vector admin 2
Vector admin 3
Vector admin 4
Tumor harvest
Flow for GFP+ Tumor cells
Imaging once a week
RV-GFP+ tumor cells
ns
% GFP+/ hub2M+
control
GEN1017
GEN3117

Fig. 9 Superior transduction by GEN3117 over GEN1017 in human ovarian cancer model in immunocompromised mice
SKOV3ip-fLuc2 cells were intraperitoneally injected into immunodeficient mice. Five days post injection, GEN3117, GEN1017, or control media was injected intraperitoneally 4 times every 5 days to n=6 mice per group. For each injection, 3e5 TU of each vector was injected per mouse per day. Tumor growth was tracked by bioluminescence imaging, and overall growth was monitored by animal weight (no difference between groups, data not shown). On day 23, animals were sacrificed, and tumors were harvested from the abdominal cavity. Collected cells were dissociated and subjected to flow cytometry, using an antibody against anti-human β2 macroglobulin (hu β2M) to identify the SKOV3ip-fLuc2 human tumor cells. The population of GFP-positive human tumor cells (GFP+/hu β2M+) was significantly higher in the GEN3117 group than in control and GEN1017 groups.

CONCLUSIONS

Our newly developed GVO vector platform pseudotyped with optimized SB-based targeting envelopes showed target cell specificity and increased transduction efficiency. In particular, GVO vectors displaying anti-HER2 scFv or DB in the E2 domain of SB env both efficiently transduced HER2-positive SKBR3 breast cancer and SKOV3 ovarian cancer cells, with anti-HER2 DB-targeted vectors showing higher transduction efficiency compared to scFv-targeted vectors. In contrast, HER2-negative MDA-MB-468 breast cancer cells were not transduced, unless engineered to overexpress HER2. Furin cleavage seems to play an essential role in vector transduction; the backbone envelope structure with the furin site showed greatly improved transduction (However, the furin site in the antigen-binding domain greatly diminished the transduction and needed to be eliminated to restore transduction). Our initial *in vivo* study indicates higher transduction by anti-HER2 DB-targeted vector compared to the amphotropic control vector administered at the same infectious titer. Several additional targeting vectors have been also developed and their transduction specificity and efficiency are being confirmed.

ACKNOWLEDGMENT

Special thanks to the R&D, Pre-Clinical and PSE teams at GenVivo, Inc. and the team at UCSF.

CONTACT

Makoto Sato, PhD: GenVivo, Inc., San Marino, CA, msato@genvivoinc.com, 626-441-6695
GenVivo Inc. is a clinical-stage gene therapy company headquartered in San Marino, CA, currently advancing a cancer immunotherapy enveloped vector product candidate.