Serial assessment of GEN2, a retroviral vector for gene transfer of an optimized thymidine kinase and GM-CSF, for genomic integration into peripheral blood mononuclear cells (PBMCs) in a Phase 1 trial in adult patients with solid tumors

Alison L. Hannah[1], Noriyuki Kasahara[2], Li Liu[1], Allen Wang[1], Joe McNulty[1], Robert G. Johnson[1]
[1]GenVivo Inc., San Marino, CA; [2]Neurological Surgery, University of California San Francisco, San Francisco, CA

AACR 2024
Abstract
7250 / 17

Introduction

GEN2 is a non-replicating retroviral gene transfer vector encoding an enhanced herpes simplex virus thymidine kinase (HSV-eTK), which activates the pro-drug ganciclovir (GCV), resulting in chain termination during DNA replication, triggering cancer cell apoptosis and release of neoantigens (NA). GEN2 also encodes human Granulocyte Macrophage-Colony Stimulating Factor (hGM-CSF), which augments antitumor immunity by enhancing NA presentation to immune effector cells.

Linear Sequence Map of GEN2 Vector Genome

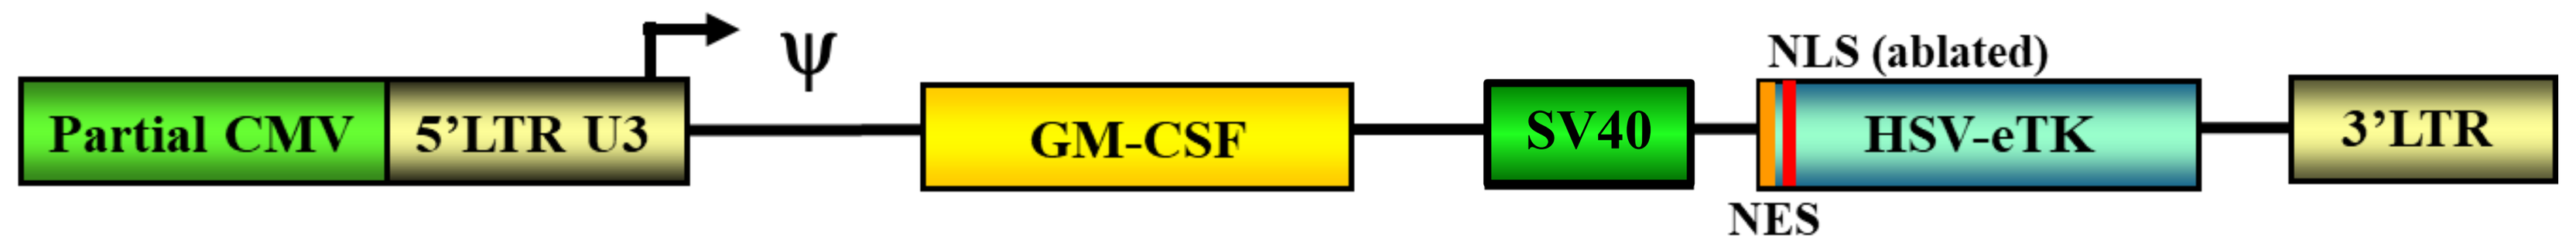
Partial CMV
5'LTR U3
Ψ
GM-CSF
SV40
NLS (ablated)
HSV-eTK
NES
3'LTR

GEN2 Vector Particle

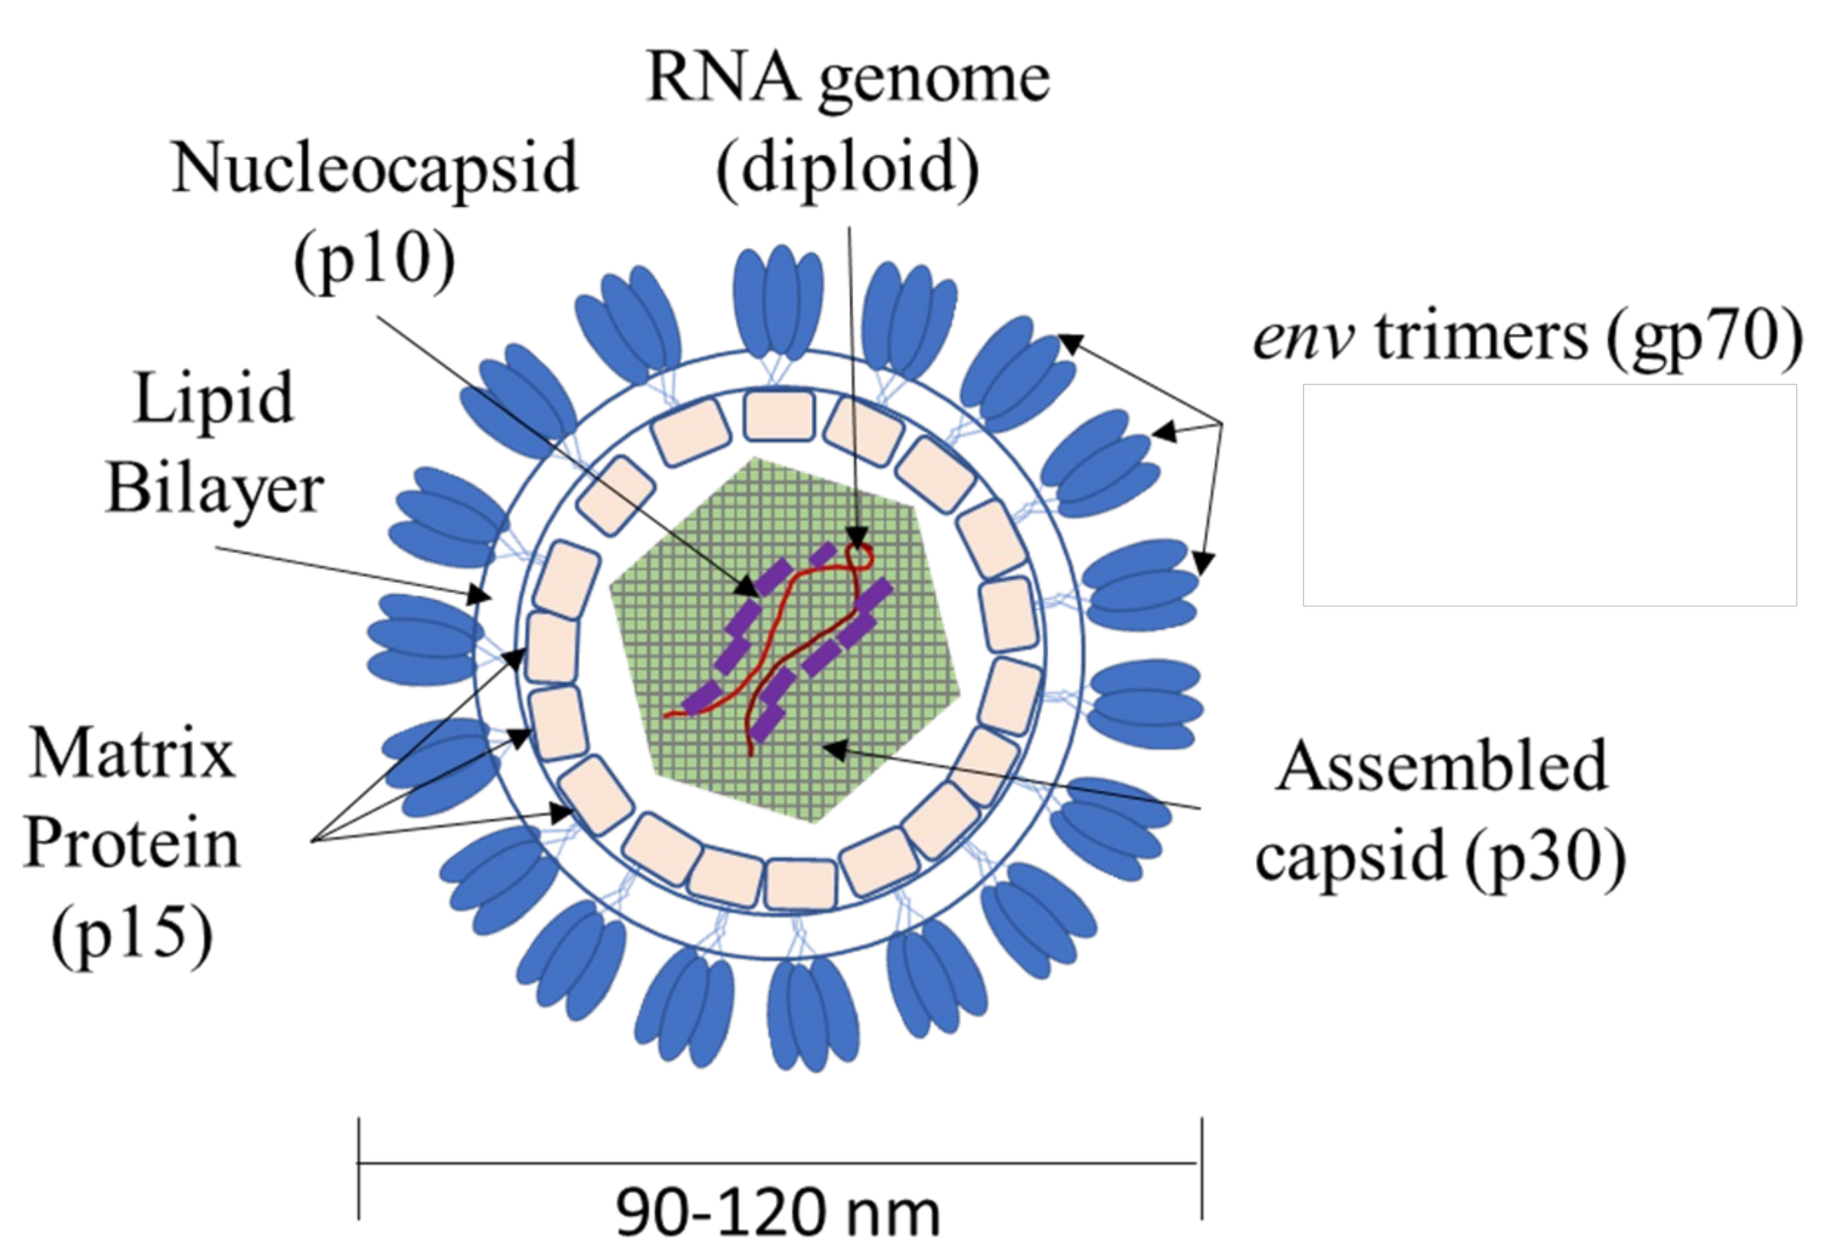
RNA genome (diploid)
Nucleocapsid (p10)
Lipid Bilayer
env trimers (gp70)
Matrix Protein (p15)
Assembled capsid (p30)
90-120 nm

CMV: partial cytomegalovirus promoter
LTR: retroviral long terminal repeat
Ψ (Psi): retroviral packaging signal
SV40: simian virus 40 promoter
NES: nuclear export signal
NLS: nuclear localization signal
p10, p15, p30, gp70: Proteins present in the GEN2 vector particle, but encoding gene sequences are absent from the vector genome

GEN2 in combination with valganciclovir (an oral formulation of GCV) was evaluated over 12 escalating GEN2 dose levels (6.4×10^6 – 3.4×10^9 Transducing Units (TU)) in a first-in-human Phase 1 clinical trial (NCT04313868) in n=56 adult patients with advanced solid tumors. Serial assessment of peripheral blood mononuclear cells (PBMCs) to detect GEN2 integration and any potential replication-competent retrovirus (RCR) was performed by multiplex digital droplet polymerase chain reaction (ddPCR) of PBMC genomic DNA.

Methods

Patients were enrolled into 3 cohorts employing different routes of administration (IV: intravenous, HAI: hepatic artery infusion, IT: intratumoral), and patient PBMCs were acquired for GEN2 integration and RCR testing, per the schematic below:

GEN2 & Valganciclovir (VGCV) Dose Cycles by Administration Route

GEN2 Administration
IV: Day 1/2/3
HAI: Day 1/2
IT: Day 1

→ **VGCV Administration**
Daily on Days 8-12

→ **Drug Holiday**
Day 13 to Day 21
Then begin next cycle

Patient PBMC Sampling Schedule for GEN2 Integration & RCR

Eligibility Screening & Patient Enrollment → First Two Dose Cycles (Cycle 1, Dose 1 Pre-infusion; Cycle 2, Dose 1 Post-infusion) → Post-infusion sampling every 6 months until death or withdrawal of consent

Results

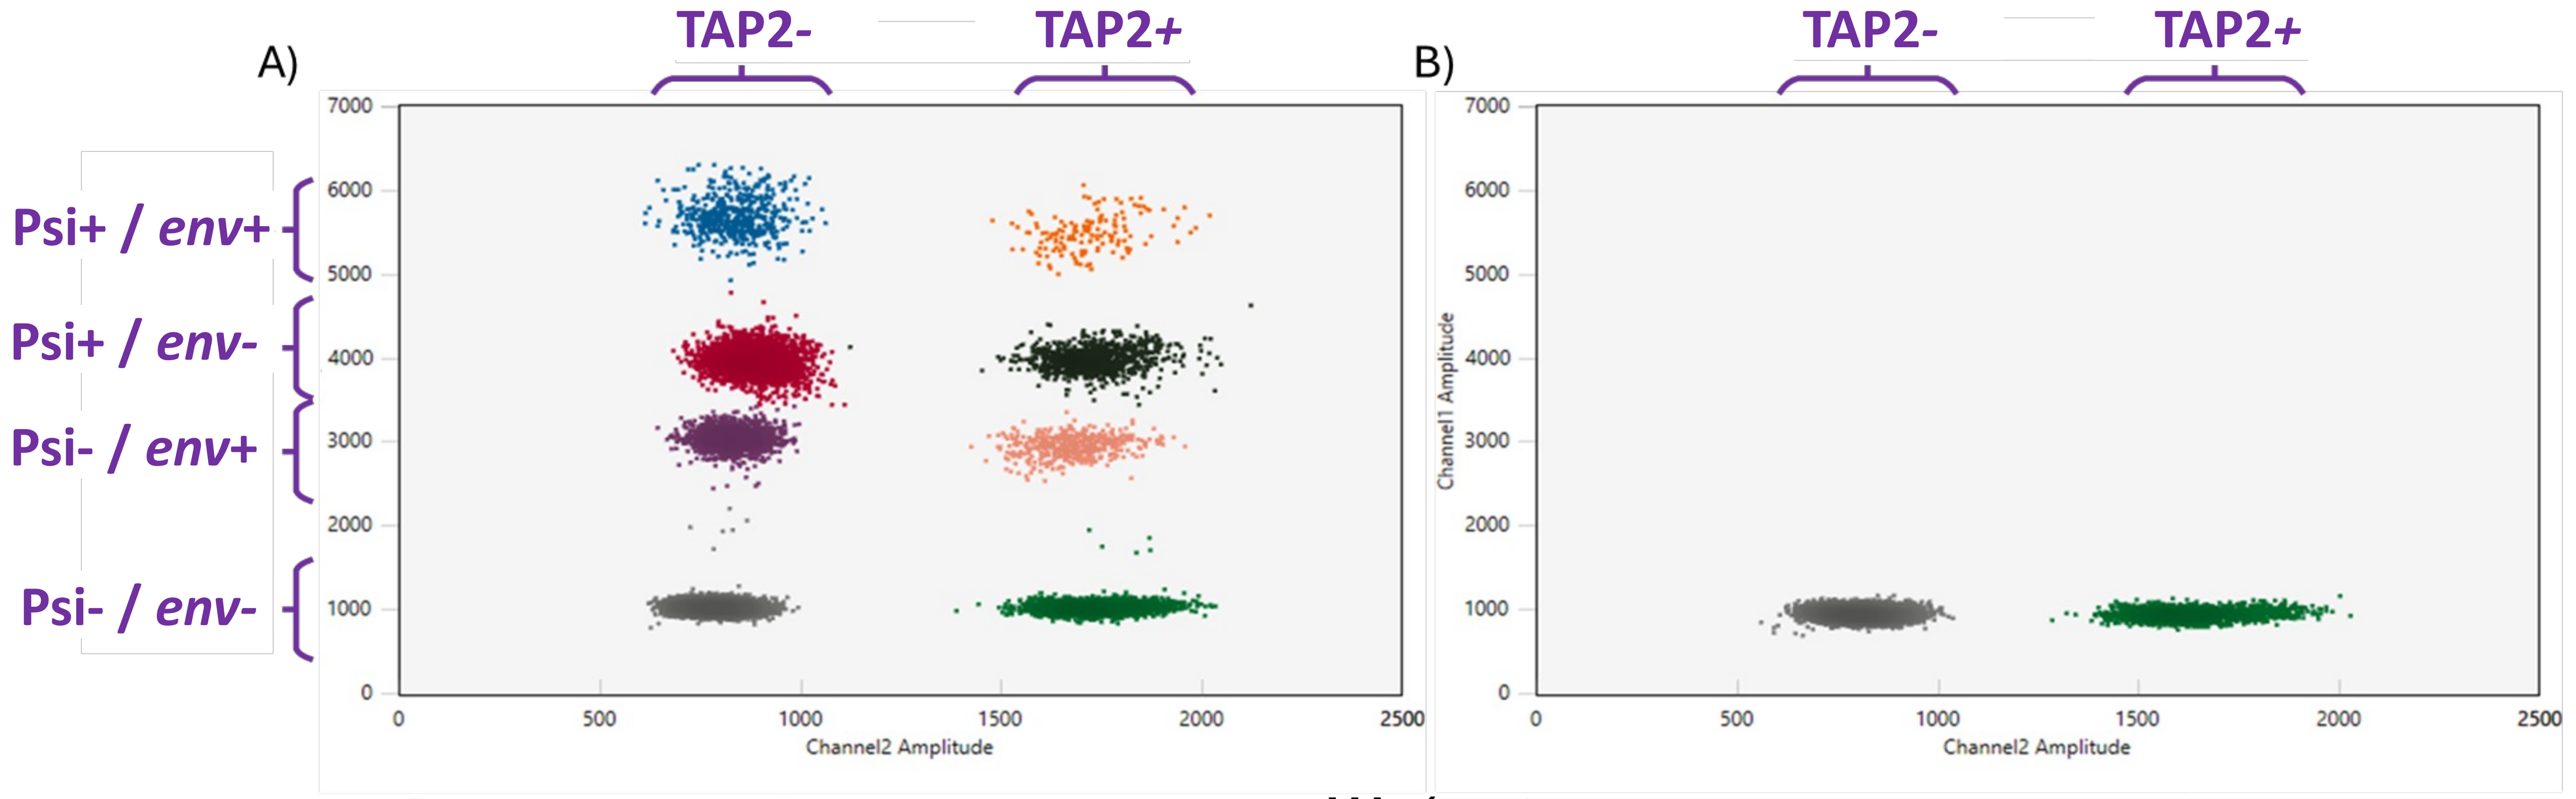
A)
B)
TAP2-
TAP2+
Psi+ / env+
Psi+ / env-
Psi- / env+
Psi- / env-
Channel1 Amplitude
Channel2 Amplitude

Detection of DNA encoding GEN2 Vector Ψ (Psi) and *env* sequences by ddPCR is shown as 2-D plots for: A) Positive control cell sample engineered with Psi and *env* genes; B) representative patient PBMC sample. The right column in both A) and B) panels show detection of the TAP2 housekeeping gene as an internal assay control. The presence of Psi sequences would indicate stable integration of the GEN2 vector, and *env* sequence detection would indicate the presence of RCR. All results for GEN2 vector integration and RCR were below the limit of quantitation, indicating that mature PBMCs are resistant to vector transduction and there is no evidence of RCR.

Discussion

Clinical evaluation of GEN2 is on-going in adult patients with solid tumors. In all patient samples tested to date, regardless of dose level or route of administration, there has been no evidence of GEN2 integration into PBMCs or of RCR reversion.

Per the 2020 FDA Guidance for Industry (Long Term Follow-Up After Administration of Human Gene Therapy Products), serial assessment of vector integration is mandated throughout a patient's participation in a gene therapy clinical trial, and for up to 15 years following the last dose.

These guidance documents were originally conceived when gene therapy was developed to treat rare hereditary diseases in pediatric patients; however, adult patients in early-stage clinical trials for cancer typically do not have a prolonged life expectancy (BMC Cancer 2011 11:426).

Moreover, a recent review noted no evidence of RCR detected in 1,595 post-treatment PBMCs obtained from 60 clinical trials (Mol. Ther. 2023 31(3):801). Also, RCR has not been detected in any clinical lot of retroviral vector to date (Mol. Ther. Meth. & Clin. Dev. 28:28).

In the context of proper and warranted usage of patients' blood for testing, attention should be drawn to the limited expectation for a 2nd malignancy within the lifespan of this patient population, and the absence of detectable RCR in cancer patients receiving retroviral vectors over decades.

Conclusions

Our results confirm undetectable genotoxic risk of GEN2 for gene therapy of cancer at all doses tested to date. For clinical trials of gene therapy enrolling adult cancer patients, a 1-year follow-up period appears justified.

Contact
Robert G. Johnson, MD PhD
GenVivo, Inc.
San Marino CA
rjohnson@genvivoinc.com

References:
1. Long Term Follow-Up After Administration of Human Gene Therapy Products (January 2020 FDA Guidance for Industry; www.fda.gov).
2. Chau et al. Early mortality and overall survival in oncology phase I trial participants: can we improve patient selection? BMC Cancer 11, 426 (2011).
3. Cornetta et al. Replication competent retrovirus testing (RCR) in the National Gene Vector Biorepository. Mol Ther 2023 31(3):801
4. Cornetta et al. Meeting FDA Guidance recommendations for replication-competent virus and insertional oncogenesis testing. Mol. Ther. Meth. & Clin. Dev. 2023 28:28

© POSTER TEMPLATE BY GENIGRAPHICS® 1.800.790.4001 WWW.GENIGRAPHICS.COM